APPLICATION OF NEXT GENERATION *IN VITRO* APPROACHES FOR THE ASSESSMENT OF E-CIGARETTES

Marianna Gaca, **Frazer Lowe, Emmanuel Minet, David Throne and Damien Breheny**

British American Tobacco (Investments) Ltd, Southampton, SO15 8TL, UK

Correspondence: marianna_gaca @bat.com

Introduction

Electronic cigarette (e-cigarette) use has increased at a rapid pace over the last decade. As these products continue to evolve to meet consumer demand, there is an increased requirement for the toxicological evaluation of e-cigarettes and e-liquids.

Using a series of classical and next generation *in vitro* test methods, we have assessed the toxicological and biological effects of a prototype e-cigarette compared to a commercially a reference cigarette (3R4F or 1R6F).

Materials and Methods

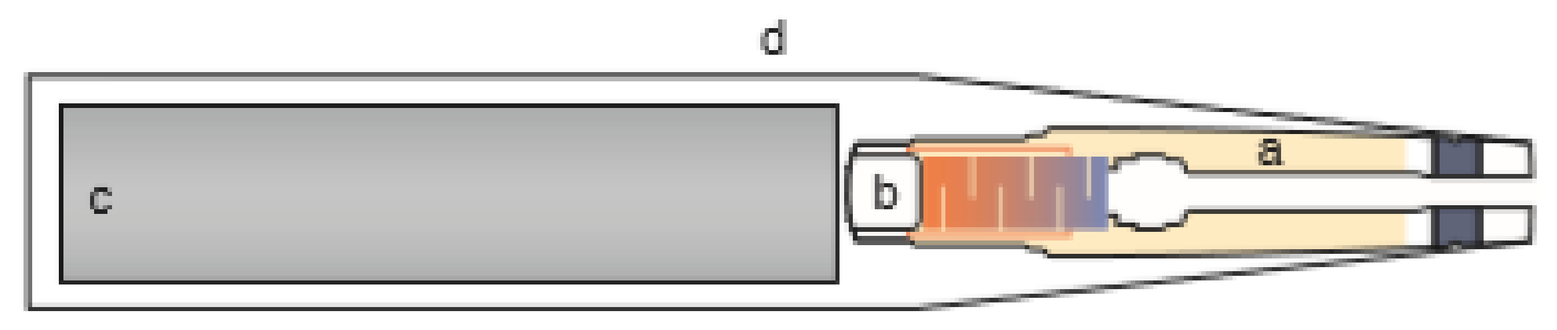

d
c
b
a

Schematic drawing of novel prototype e-cigarette. a) e-liquid; b) distiller technology; c) electronics and battery; d) device body.

Test matrix preparation

Health Canada Intense for 3R4F/1R6F reference cigarette and CORESTA Recommended Method No81 for prototype e-cigarette puffing regimens were used to generate three different test matrices for *in vitro* assessment:

- Total Particulate Matter (TPM) – trapped on a Cambridge filter pad and eluted at 24 mg/mL (3R4F) and 150 mg/mL (e-cigarette) in DMSO. TPM was diluted in cell culture medium to treat cells in submerged culture.
- Whole aerosol (WA) – cells directly exposed to aerosol at the air-liquid interface.
- Aerosol aqueous extract (AqE) – aerosol bubbled through 20 mL cell culture medium in a glass impinger. 8 puffs were used for 3R4F, while 24 puffs of e-cig were used. AqE was diluted in cell culture medium to treat cells in submerged culture.

Classical regulatory toxicology

Ames bacterial reverse mutation assay

TPMs were tested using five *S. typhimurium strains:* TA98, TA100, TA1535, TA1537 and TA102 + metabolic activation (S9) following OECD 471. For WA exposures, *S. typhimurium* tester strains TA98, TA100, TA1535, TA97 and TA102 using a modified air-agar methodology.

***In vitro* micronucleus (IVMN)**

TPMs were assessed following OECD 487, with short 3hrs exposures (± S9) and longer 24hrs -S9 exposures

Mouse lymphoma assay (MLA)

TPMs were assessed following OECD 490, with short 3hrs +S9 exposures and longer 24hrs -S9 exposures.

Neutral red uptake (NRU) cytotoxicity assay

Cytotoxicity of TPMs were assessed using BALB/c 3T3 mouse fibroblasts. WA cytotoxicity was assessed using human bronchial epithelial cells (H292) exposed at the air-liquid interface (ALI).

Bhas cell transformation assay

The potential of TPMs to induce tumour development was evaluated using the Bhas 42 cell transformation assay promoter protocol.

Next generation toxicology

Multiparametric analysis using high-content screening (HCS) approaches

The Cellomics Arrayscan VTi platform was used to assess 10 endpoints including cell health markers, DNA damage and oxidative stress, in normal human bronchial epithelial cells (NHBEs) after 4hrs or 24hrs exposures to TPM.

Luciferase-based reporter gene assay to assess oxidative stress

Antioxidant response element (ARE) transcriptional activation in stably transfected H292 cells were assessed after 6hrs and 24hrs exposures to TPM.

Endothelial cell migration assay

Human umbilical vein endothelial cells (HUVEC) were grown to confluency, "wounded" using a pipette tip and exposed to various AqE for 24hrs in 24 well ImageLock plates. Cell migration was quantified by measuring the closure of the "wound" using an Incucyte time lapse video camera and software at hourly intervals.

Systems toxicology: transcriptomics and proteomics

MucilAir human 3D primary airway epithelial cultures were exposed to 1R6F or e-cigarette aerosols at the ALI. After exposure, cells were incubated for 24hrs or 48 hrs. The cell lysates used for RNA-seq transcriptomic analysis and the air-surface liquid (ASL) was collected for LC-MSMS proteomics analysis.

Results

Classical regulatory toxicology

Cytotoxicity TPM

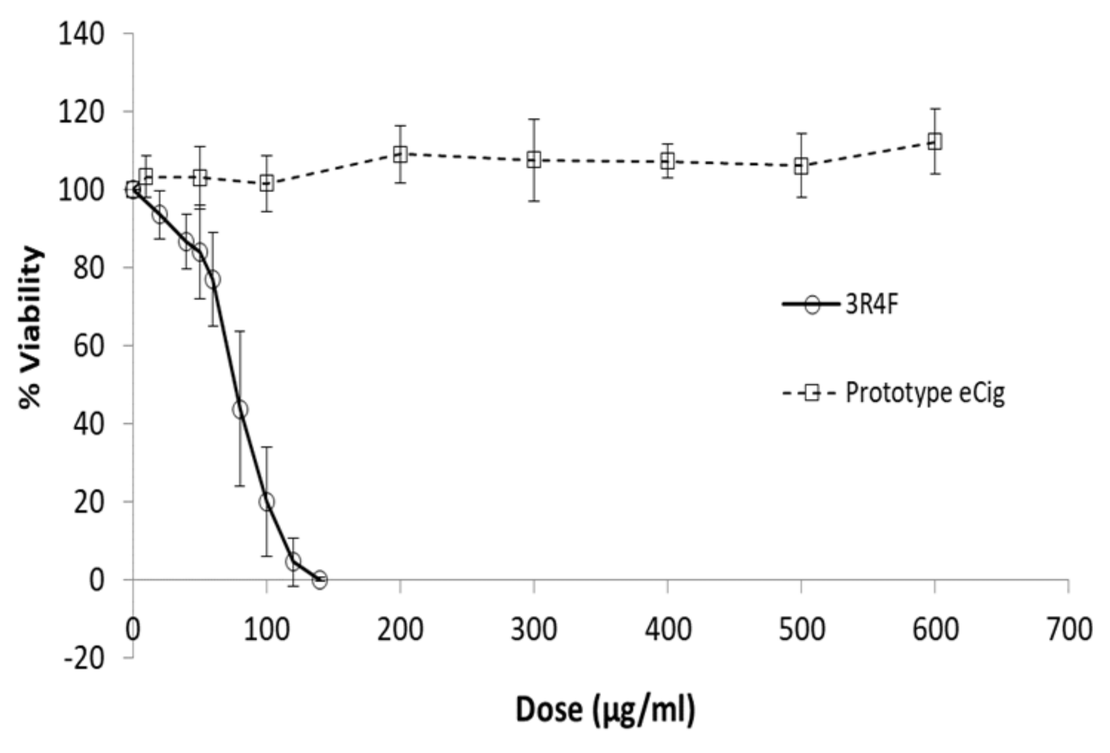

% Viability
Dose (µg/ml)
3R4F
Prototype eCig

Ames TA98 TPM

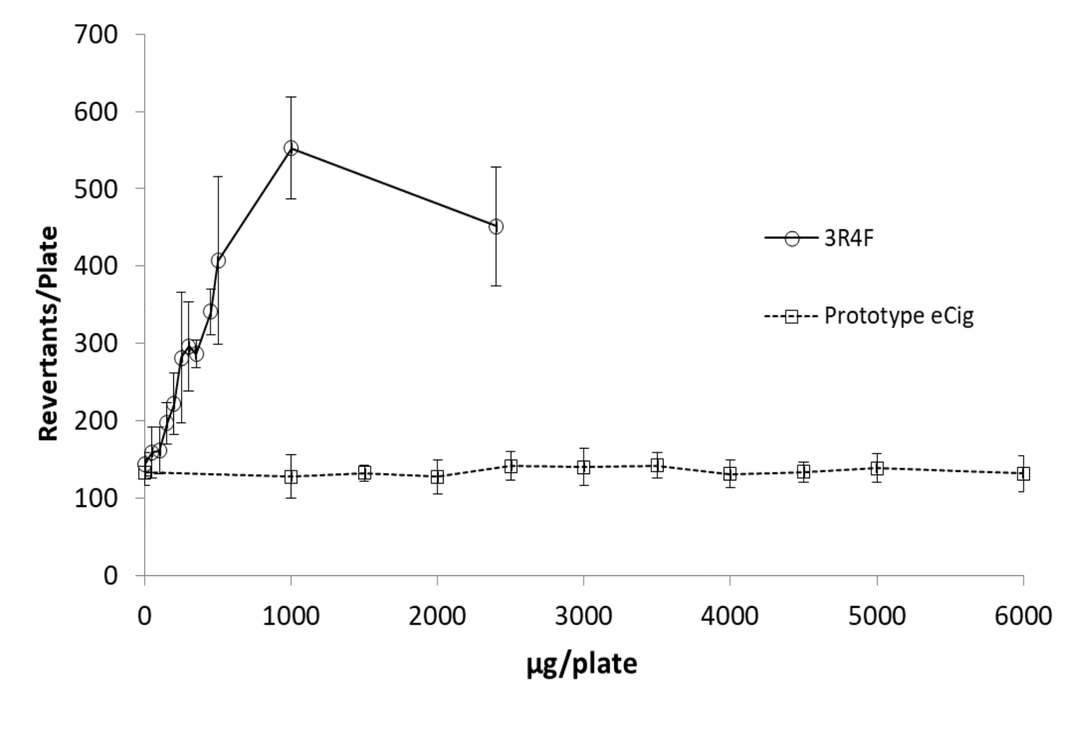

Revertants/Plate
µg/plate
3R4F
Prototype eCig

AMES TA100 TPM

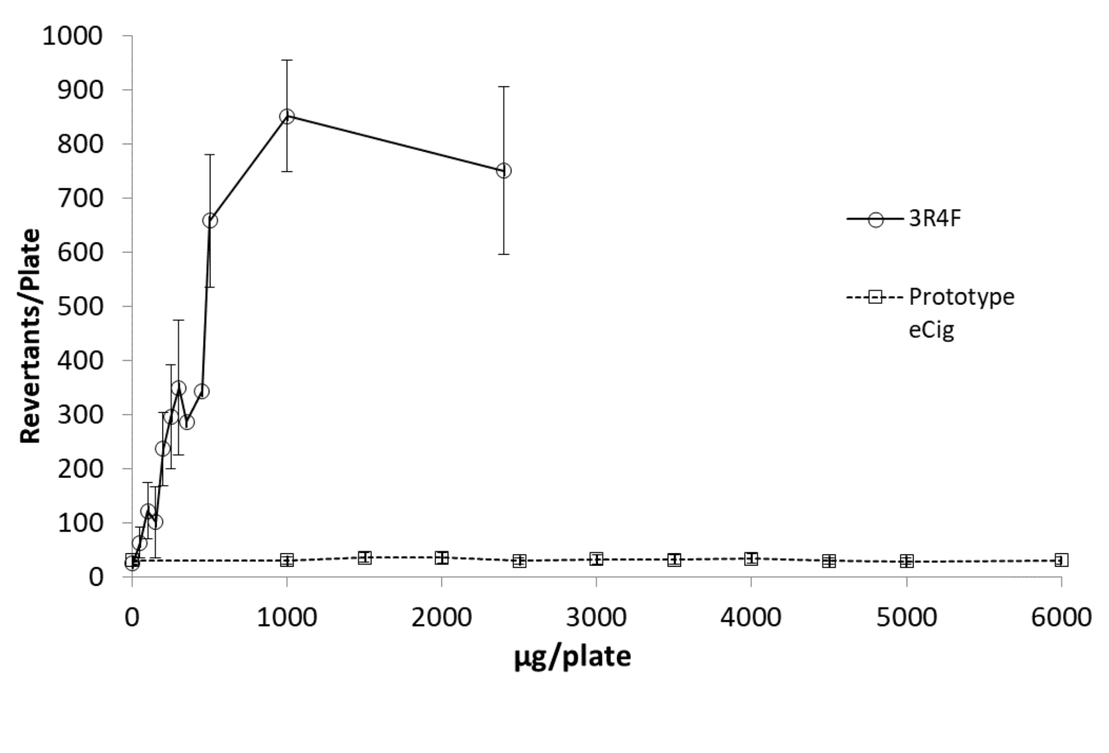

Revertants/Plate
µg/plate
3R4F
Prototype eCig

IVMN (3hrs+S9)

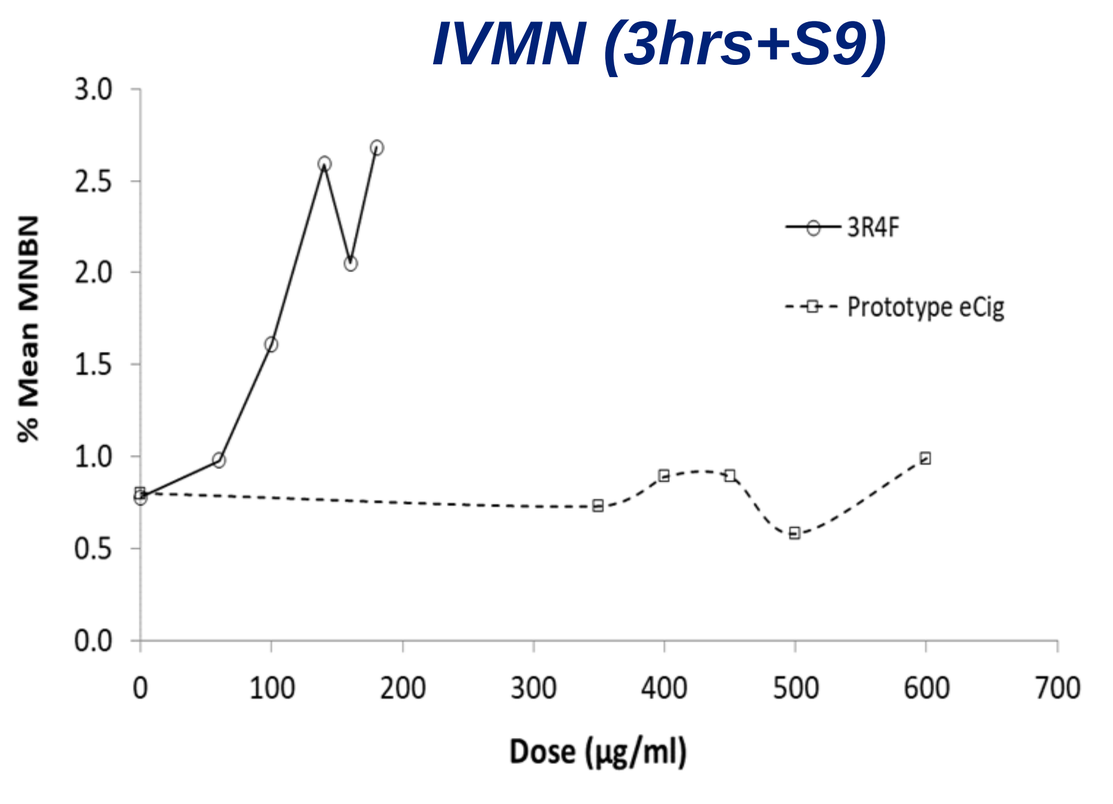

% Mean MNBN
Dose (µg/ml)
3R4F
Prototype eCig

Bhas cell transformation

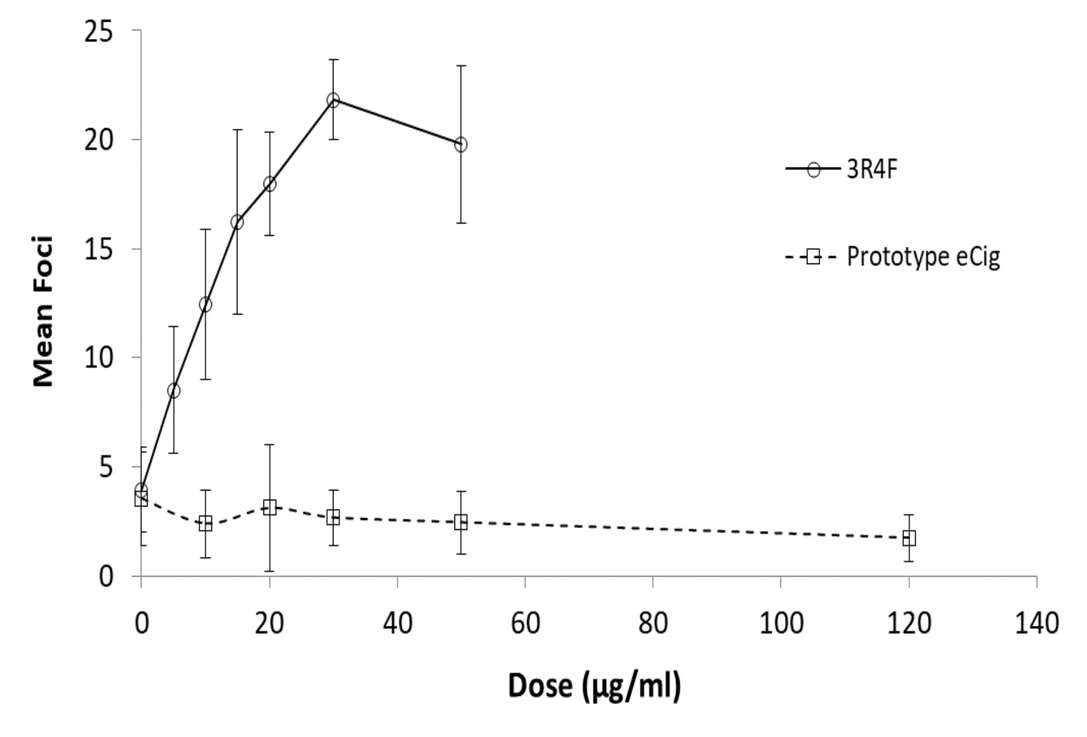

Mean Foci
Dose (µg/ml)
3R4F
Prototype eCig

MLA(3hrs +S9)

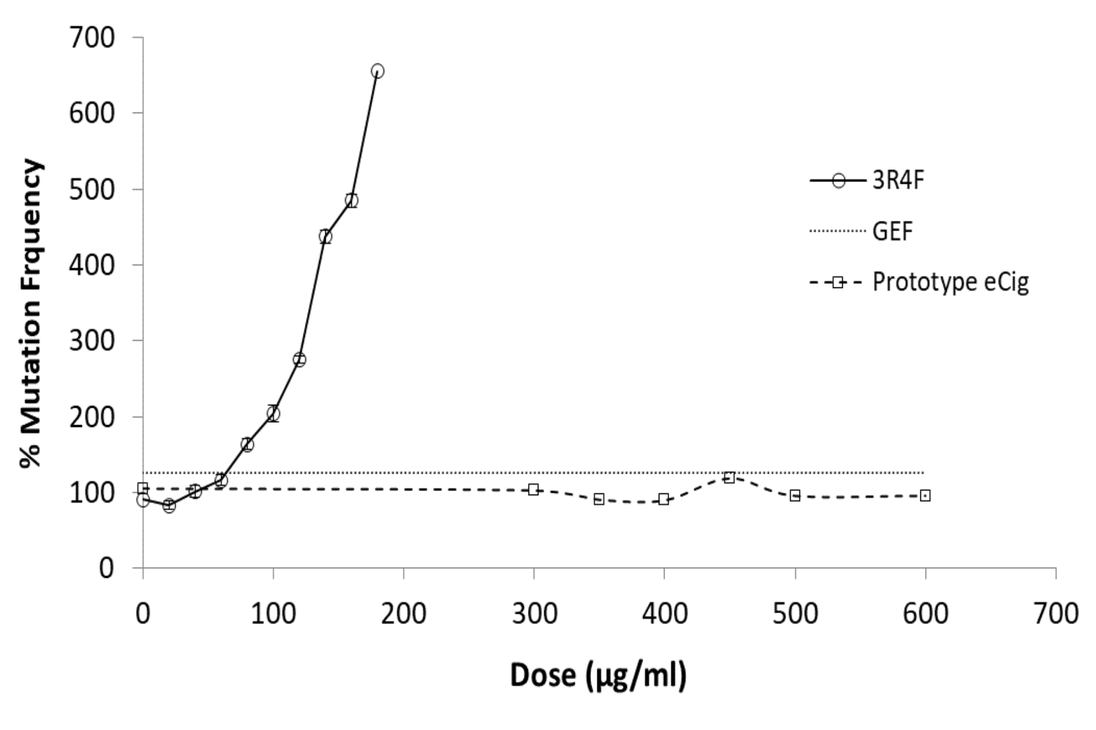

% Mutation Frquency
Dose (µg/ml)
3R4F
GEF
Prototype eCig

Ames TA98 WA

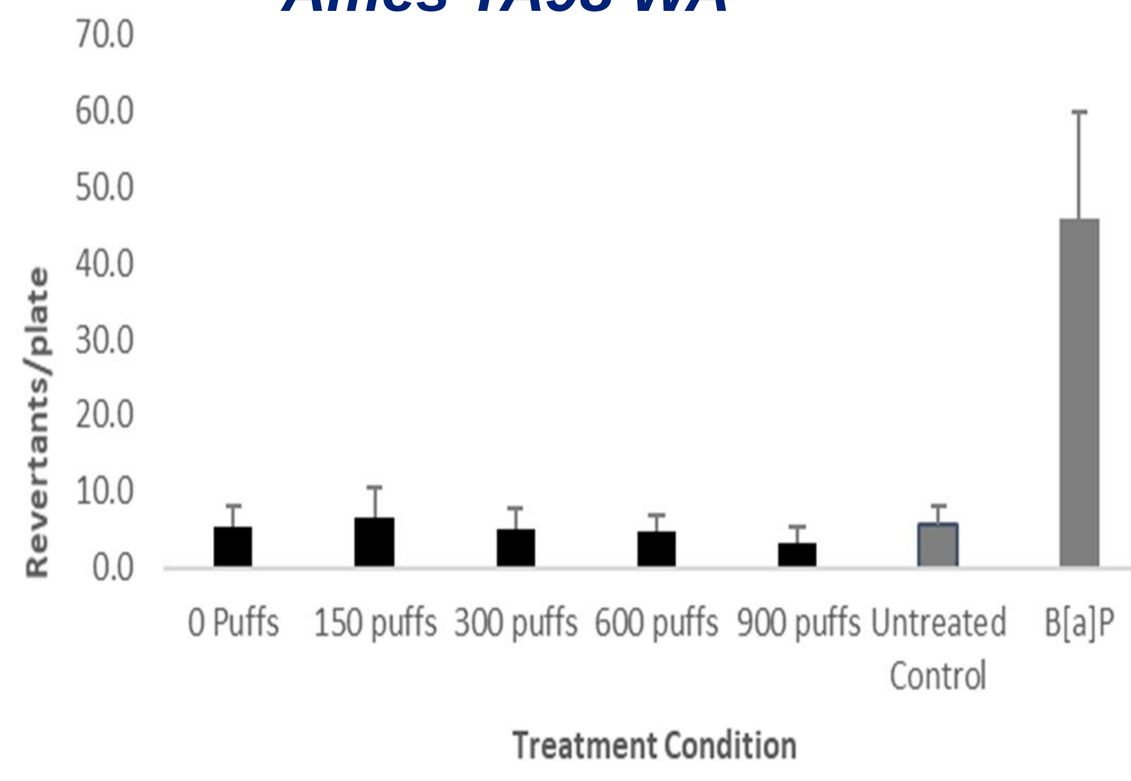

Revertants/plate
0 Puffs
150 puffs
300 puffs
600 puffs
900 puffs
Untreated Control
B[a]P
Treatment Condition

Ames TA100 WA

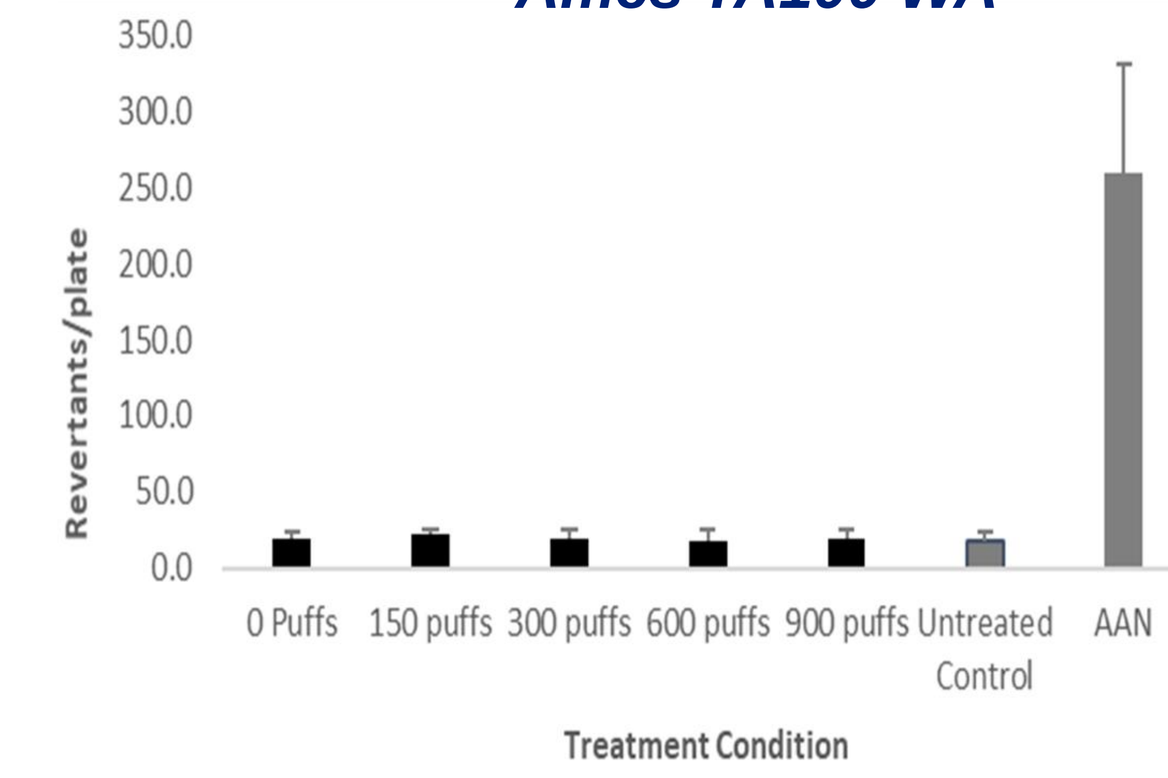

Revertants/plate
0 Puffs
150 puffs
300 puffs
600 puffs
900 puffs
Untreated Control
AAN
Treatment Condition

A battery of genetic toxicology assays including cytotoxicity, mutagenicity, clastogenicity and cell transformation were employed to test both TPM and WA matrices generated from cigarette smoke and a prototype e-cigarette.

Compared to cigarette smoke, the prototype e-cigarette showed significantly reduced activity in all assays, even at extreme doses.

Next generation toxicology

Whole aerosol cytotoxicity

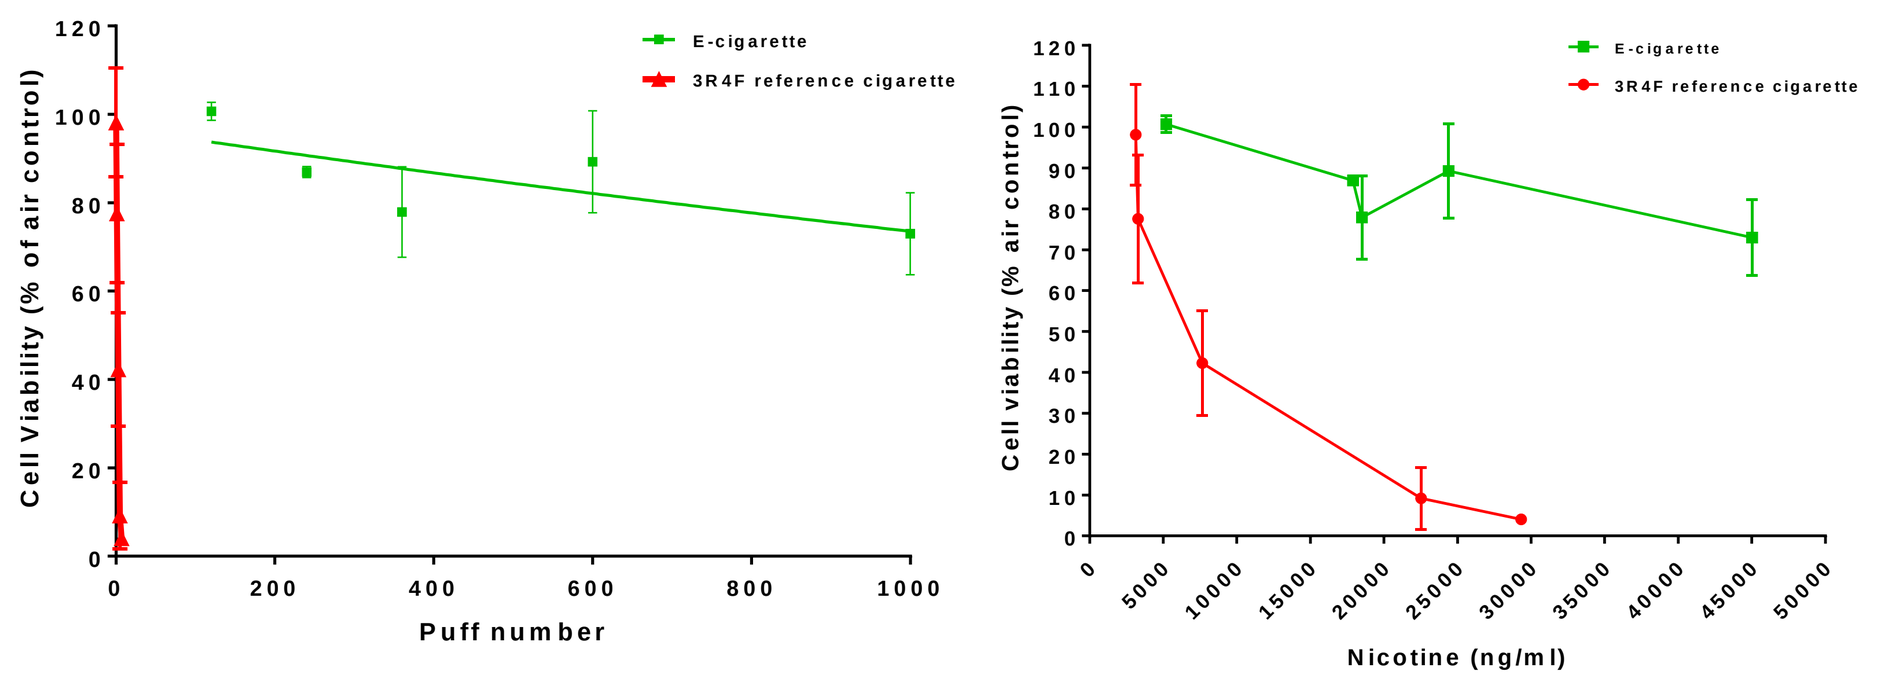

Cell Viability (% of air control)
Puff number
E-cigarette
3R4F reference cigarette
Cell viability (% air control)
Nicotine (ng/ml)

WA cytotoxicity assessment demonstrated that 3R4F produced a decrease in cell viability, resulting in complete cytotoxicity after just 7 puffs of undiluted aerosol. The novel e-cigarette induced significantly less cytotoxicity at up to 1,000 puffs and at comparable and higher levels of nicotine delivered to the cells

High content screening

HCS Endpoint	Exposure time (hrs)	3R4F	E-cigarette
Cell count	4	-	-
	24	100↓	-
Mitochondrial Mass	4	-	-
	24	200↓	-
Glutathione Content	4	200↓	-
	24	50↑ & 200↓	-
Cellular ATP	4	-	-
	24	100↓	-
DNA Damage (p-H2AX)	4	-	-
	24	200↑	-

3R4F was positive in 5 endpoints assessed using a HCS approach. In contrast, the novel prototype e-cigarette was negative for each HCS endpoint. Values are the minimum required TPM concentration (µg/mL) to elicit a ≥ 1.5-fold increase (↑) or a 30% decrease (↓) in assay signal from the 0.5% DMSO vehicle control.

Reporter gene assay

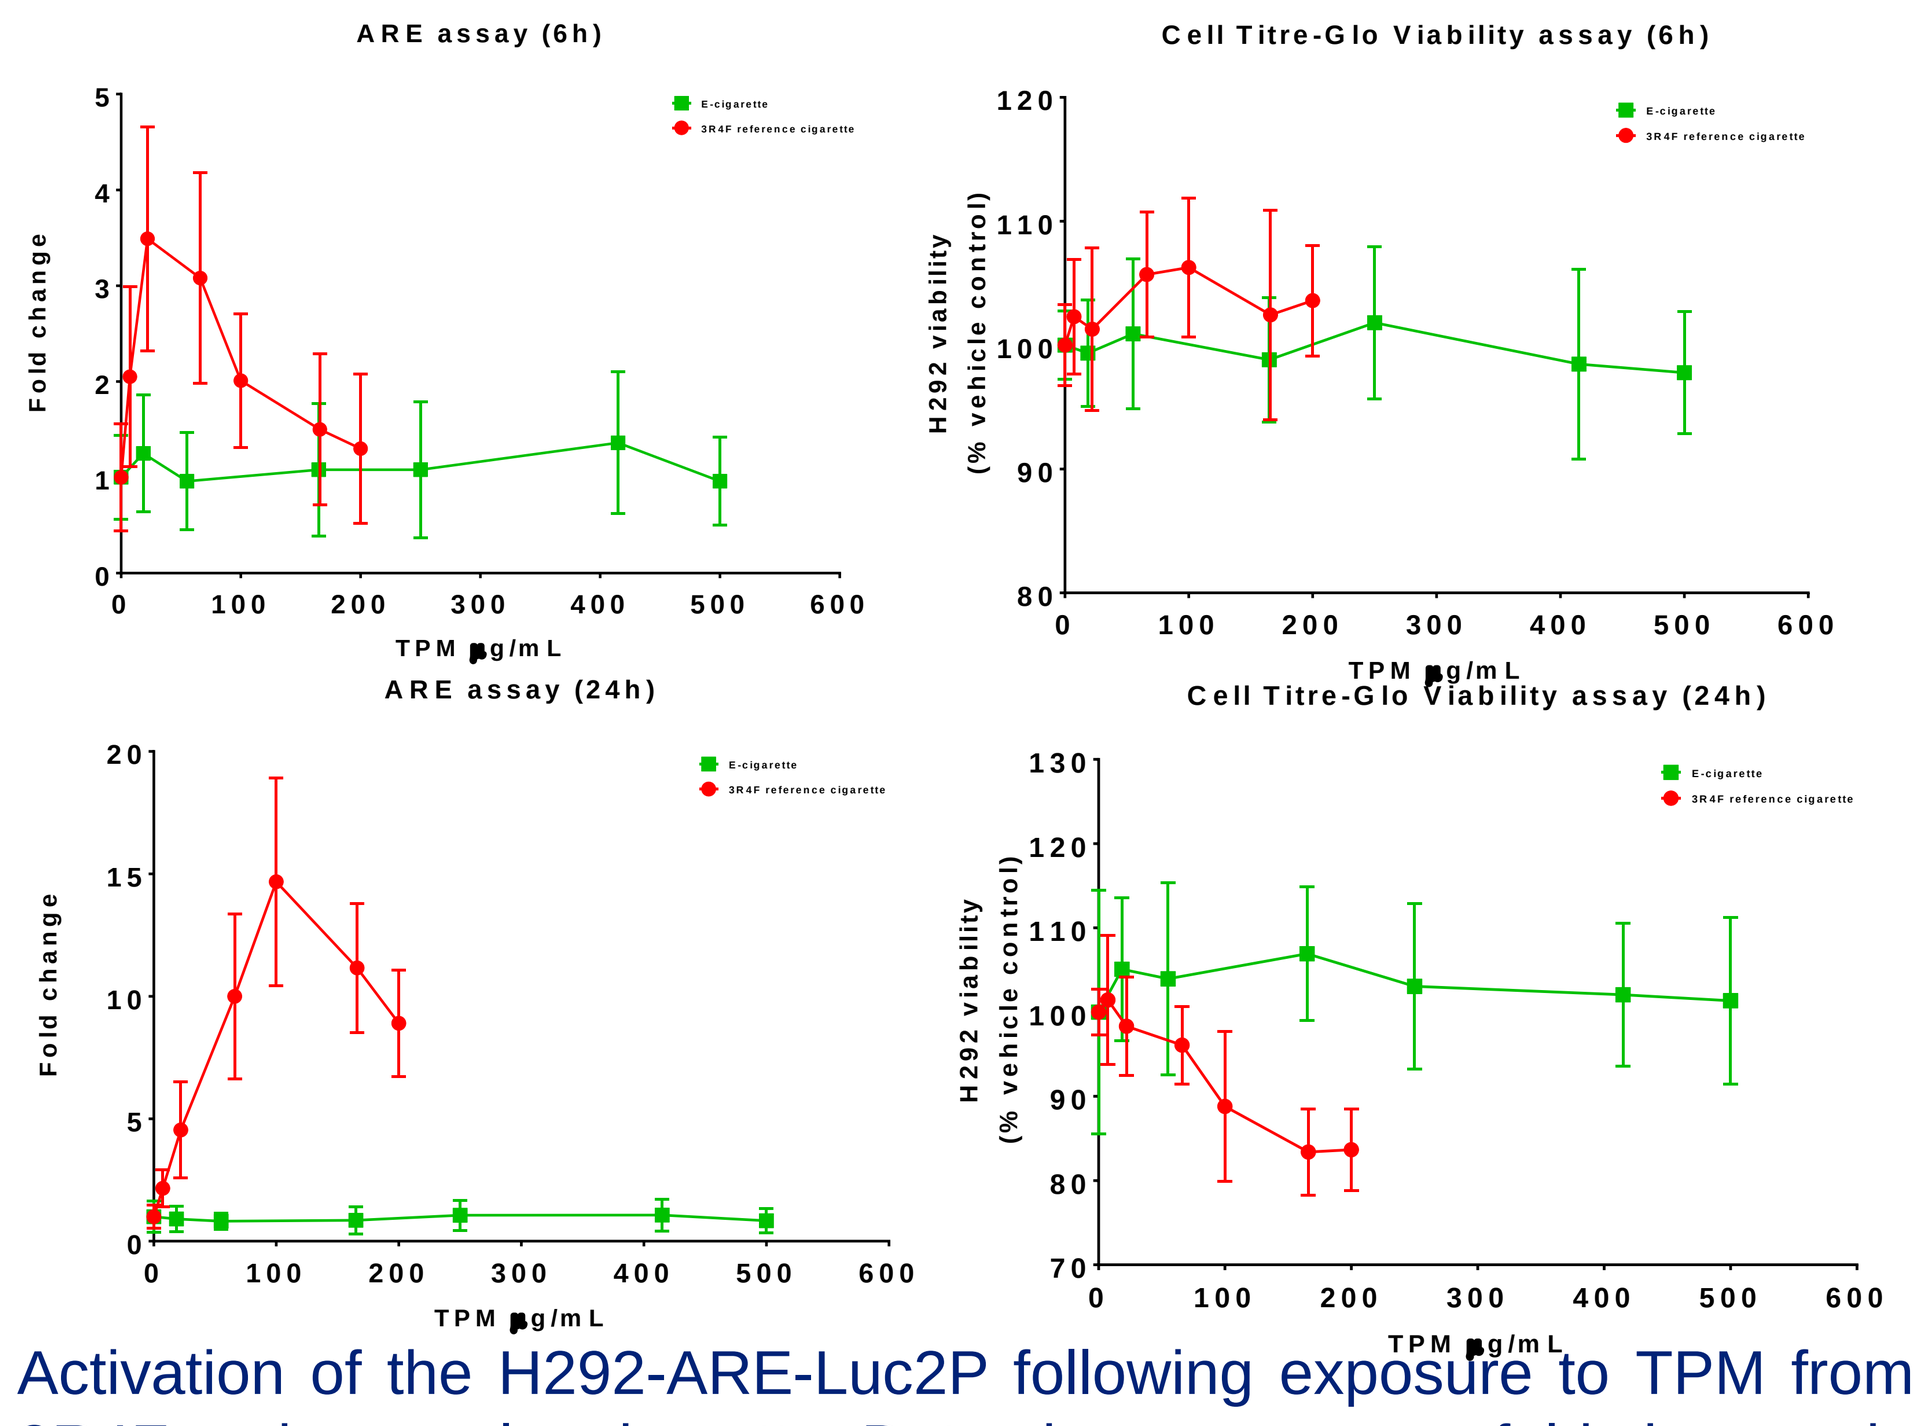

ARE assay (6h)
Cell Titre-Glo Viability assay (6h)
ARE assay (24h)
Cell Titre-Glo Viability assay (24h)
Fold change
H292 viability (% vehicle control)
TPM µg/mL
E-cigarette
3R4F reference cigarette

Activation of the H292-ARE-Luc2P following exposure to TPM from 3R4F and a novel e-cigarette. Data shown are mean fold changes in response normalized to the vehicle control (0.83% DMSO). 3R4F caused ARE activation at both time points. In contrast, the novel prototype e-cigarette was negative in the ARE assay.

Endothelial wound healing (scratch) assay

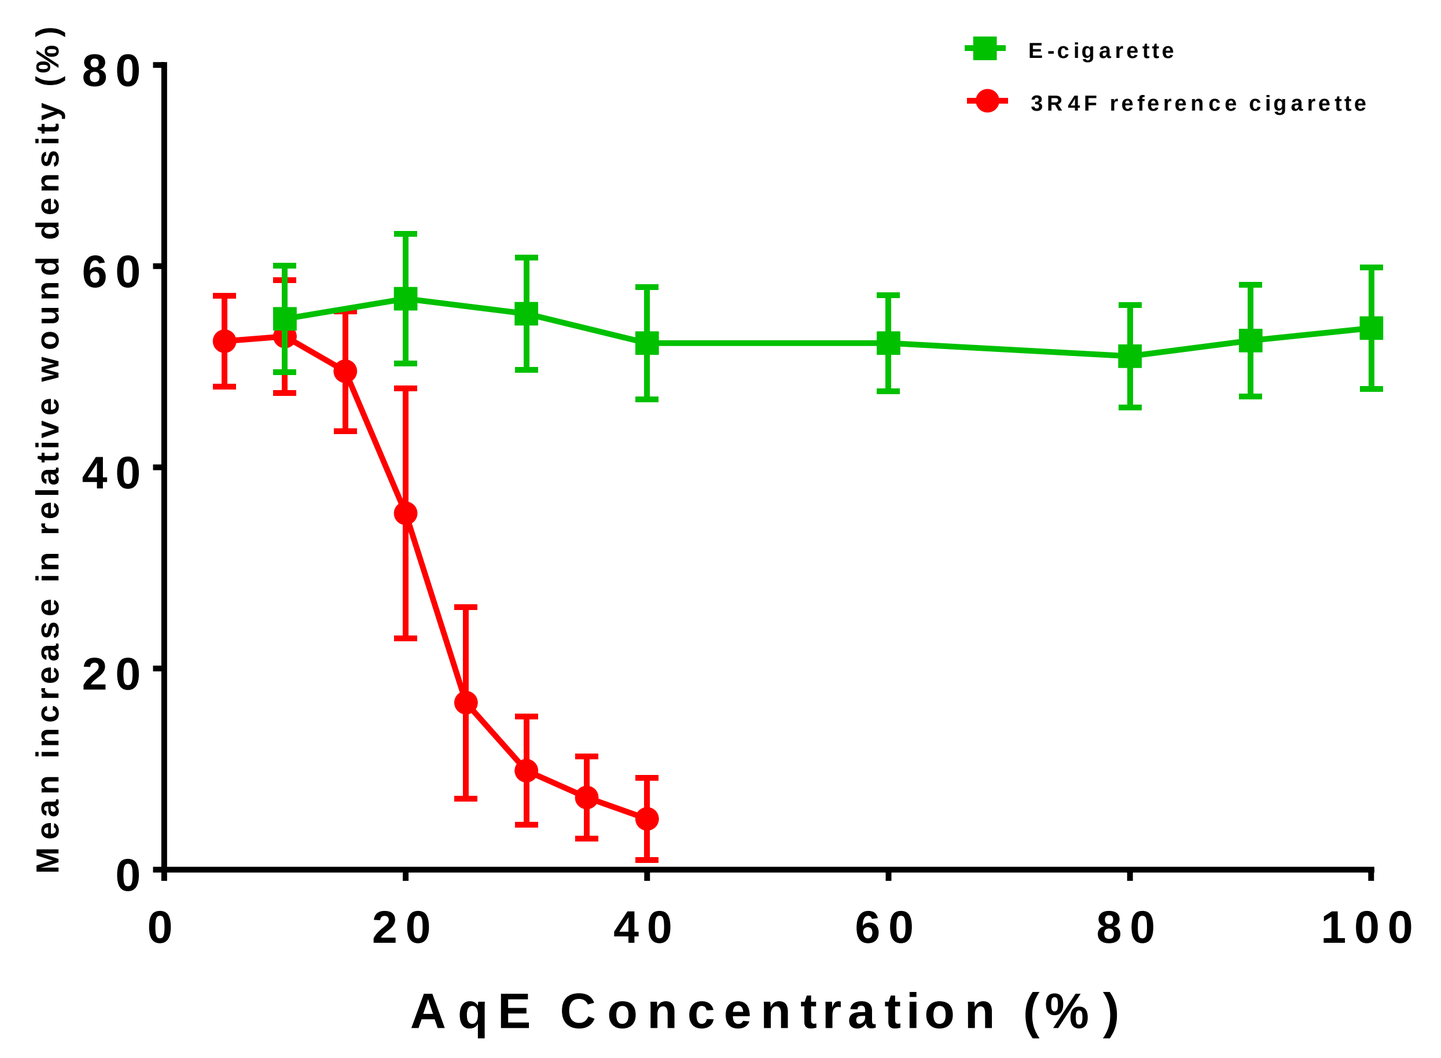

Mean increase in relative wound density (%)
AqE Concentration (%)
E-cigarette
3R4F reference cigarette

Wound healing rates (mean % increase in relative wound density) in HUVEC monolayers during 24 hour treatment with AqE from 3R4F cigarette and a prototype e-cigarette. 3R4F inhibited wound repair, in a dose-dependant manner. AqE from the novel e-cigarette did not inhibit wound repair, up to the maximum concentration tested (100% AqE).

RNA-seq differential gene expression

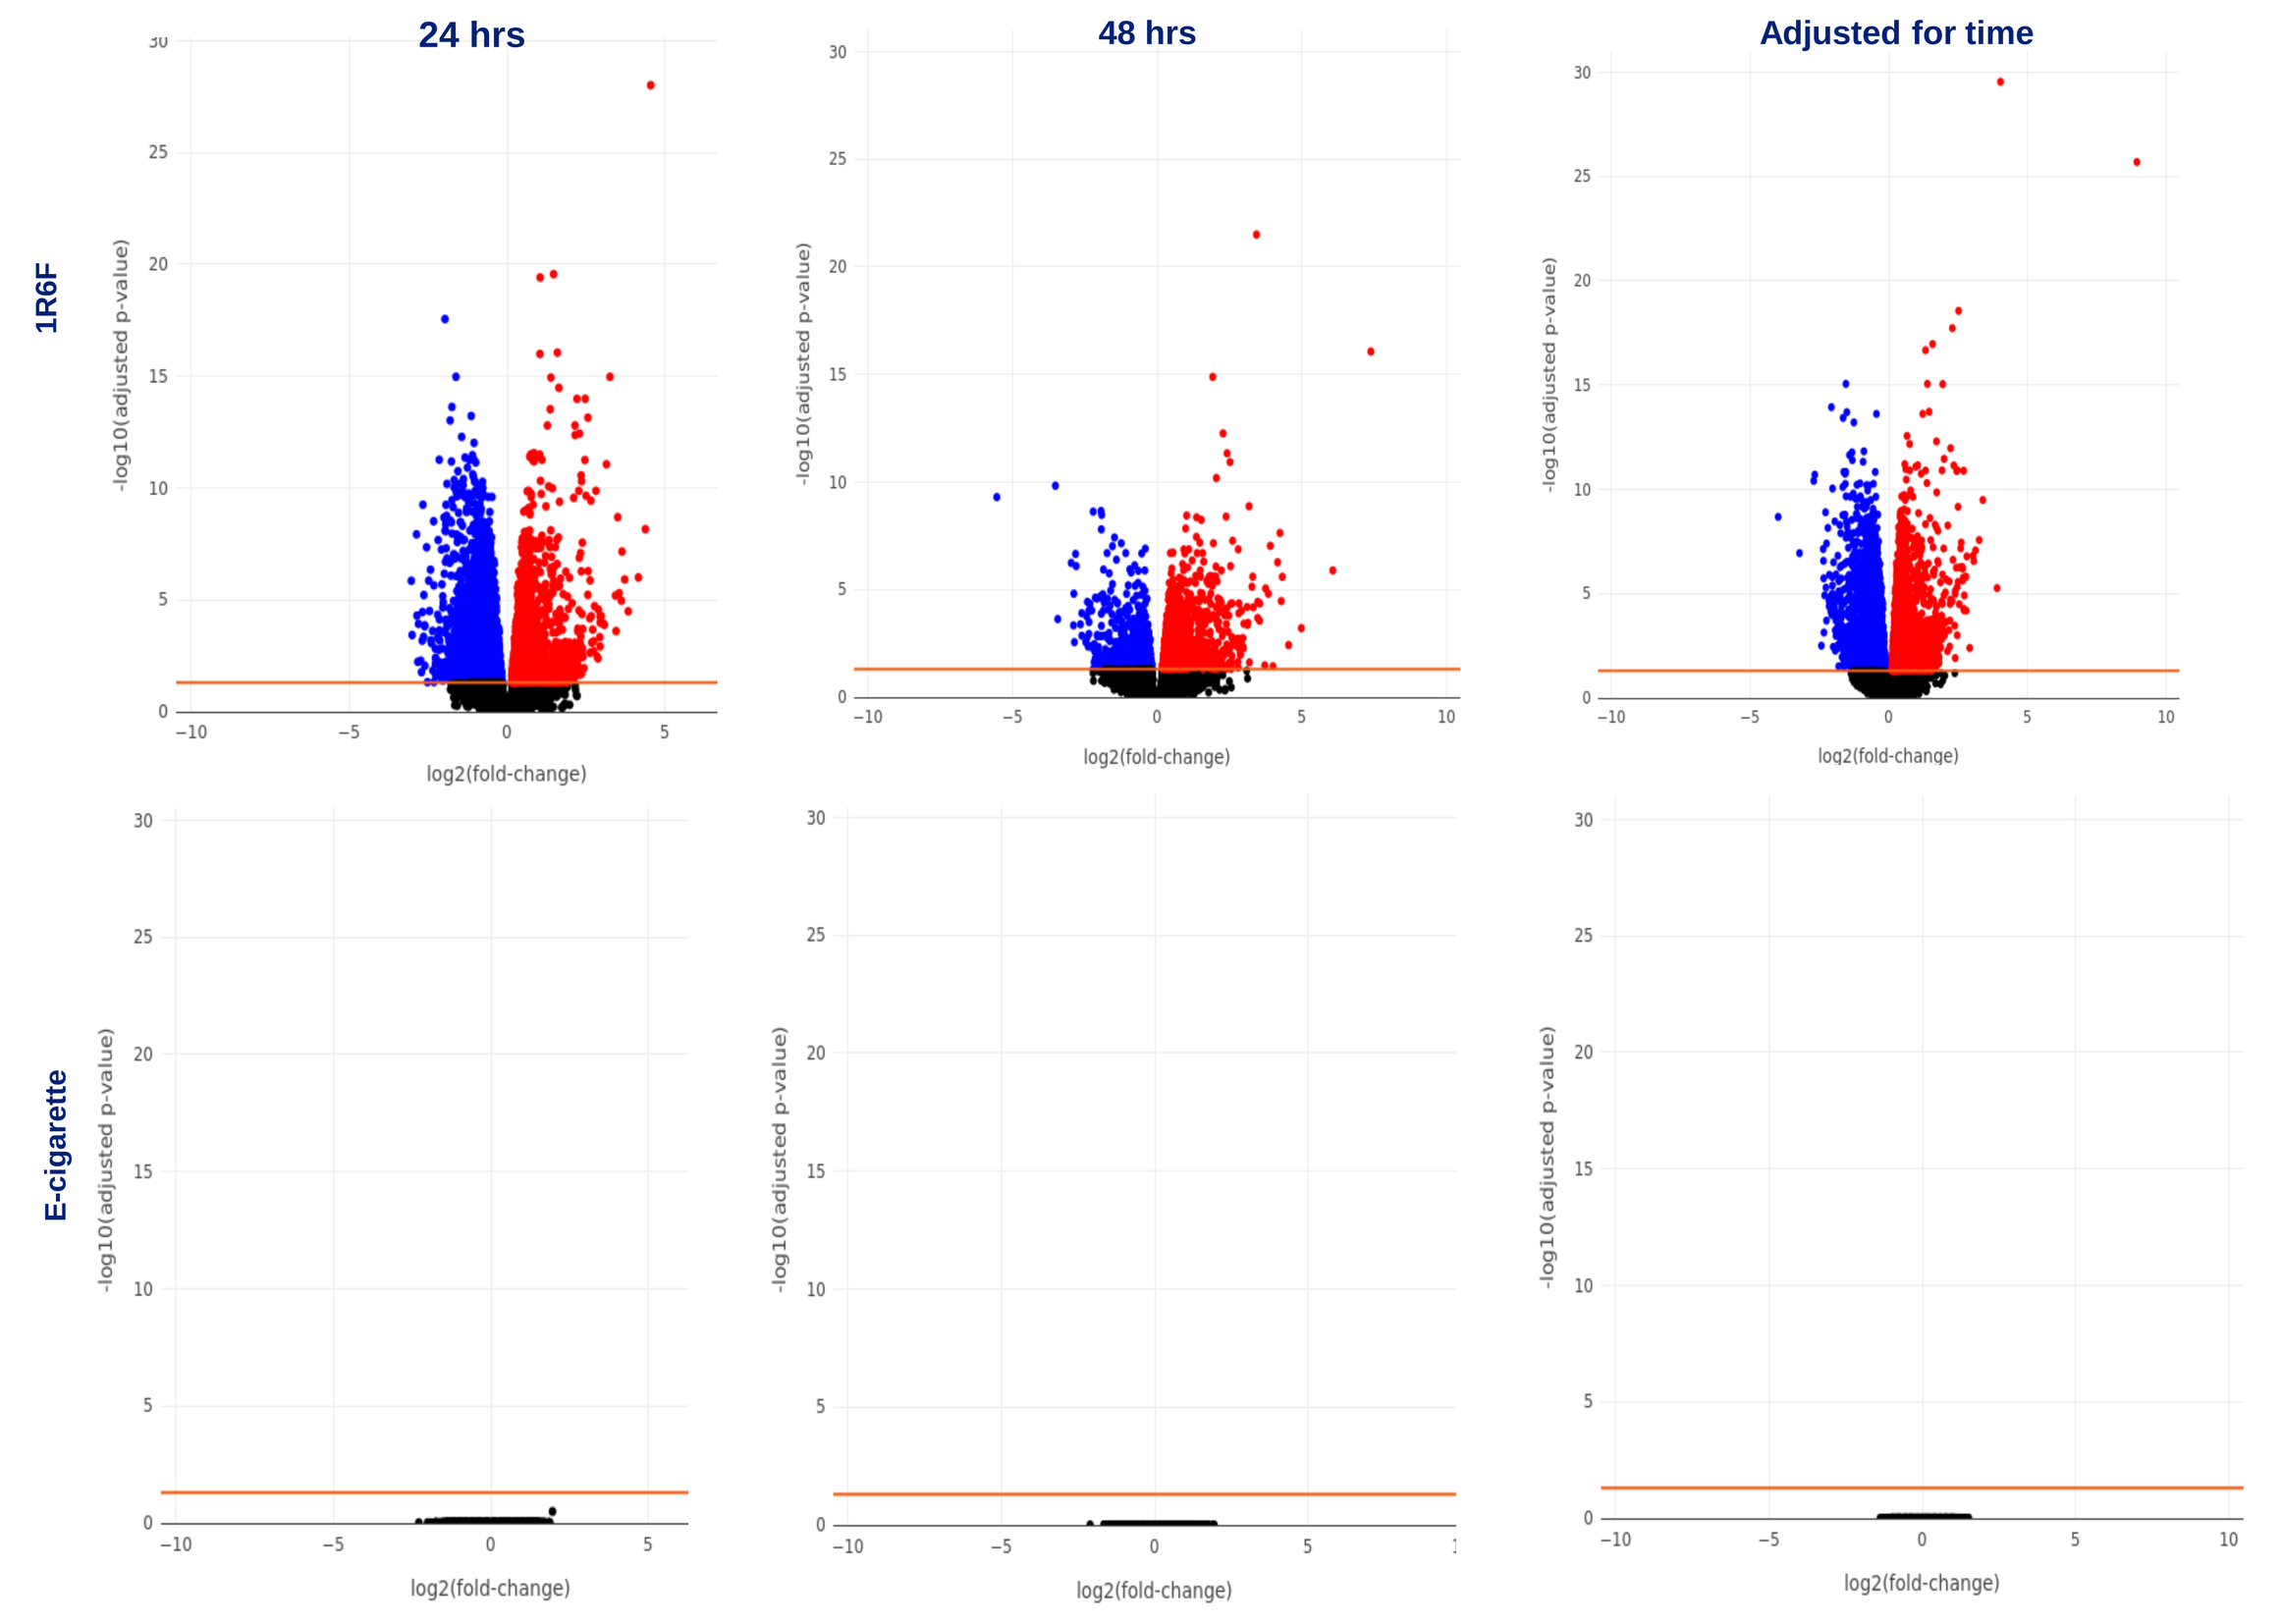

24 hrs
48 hrs
Adjusted for time
1R6F
E-cigarette
-log10(adjusted p-value)
log2(fold-change)

Transcriptomics profile of MucilAir™ after exposure to 1R6F smoke or e-cigarette aerosol**.** 1R6F exposure triggered the differential expression of 5603 genes at 24 hrs, 2180 genes at 48 hrs, and 6045 genes for the time-adjusted contrast. No differentially expressed genes were detected when comparing e-cigarette exposed RNA-seq data to the air control. Volcano plots were generated using a fold change threshold of [FC]> 1.5, and an adjusted p-value significance threshold of p<0.05.

Targetted and untargetted proteomics

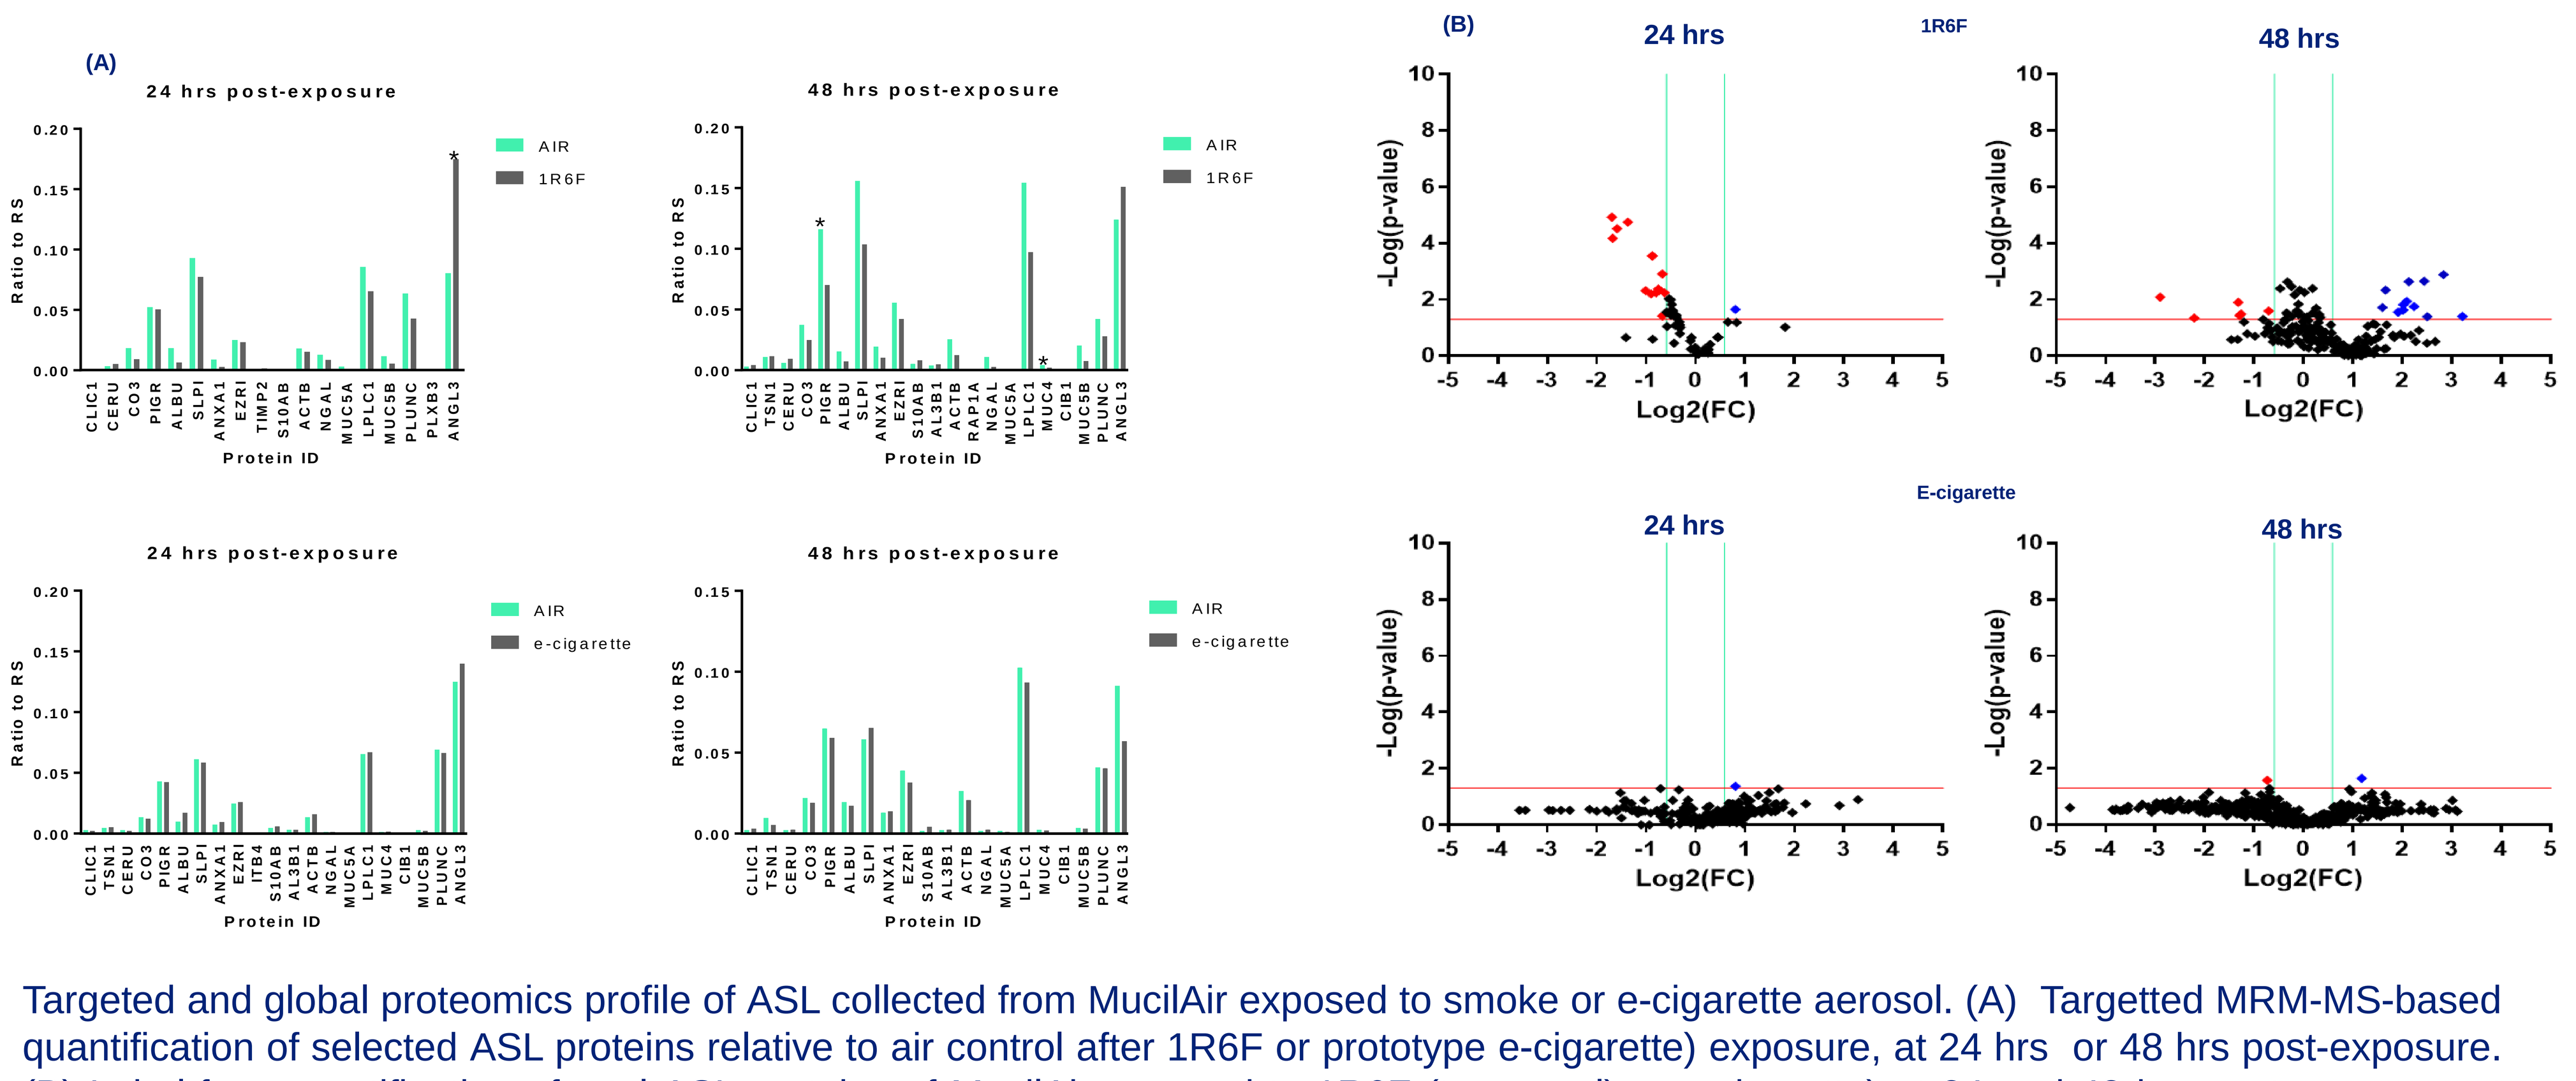

(A)
24 hrs post-exposure
48 hrs post-exposure
Ratio to RS
Protein ID
AIR
1R6F
e-cigarette
(B)
24 hrs
48 hrs
1R6F
E-cigarette
-Log(p-value)
Log2(FC)

Targeted and global proteomics profile of ASL collected from MucilAir exposed to smoke or e-cigarette aerosol. (A) Targetted MRM-MS-based quantification of selected ASL proteins relative to air control after 1R6F or prototype e-cigarette) exposure, at 24 hrs or 48 hrs post-exposure. (B) Label-free quantification of total ASL proteins of MucilAir exposed to 1R6F (top panel) or e-cigarette) at 24 and 48 hrs post-exposure at [FC] > 1.5 and p<0.05. 1R6F smoke exposure treatments with a recovery time of 24hrs and 48 hrs were found associated with a total of 16 and 17 differentially abundant proteins, respectively. At the same significance threshold, e-cigarette aerosol treatments with a recovery time of 24hrs and 48 hrs were found associated with only 1 and 2 differentially abundant proteins, respectively.

Conclusions

Compared to cigarette smoke, the prototype e-cigarette showed significantly reduced activity in all assays, even at extreme doses.

Next generation toxicological approaches can be used as part of an *in vitro* testing strategy for e-cigarettes, in concert with classical toxicological testing

The results show this novel e-cigarette has the potential to be a reduced risk product though further longer-term studies would be required to substantiate this potential.

www.bat-science.com

@BAT_Sci