In vitro dosimetric and cytotoxic assessment of a commercially available tobacco heating product (THP) versus a reference cigarette

J Adamson, T Jaunky, S Santopietro, A Terry, D Thorne, D Breheny, M Gaça
British American Tobacco, Research and Development, Southampton, SO15 8TL, UK
Correspondence: Jason_Adamson@bat.com

INTRODUCTION Tobacco Heating Product - *glo*

In the past decade, the awareness and usage of electronic cigarettes has increased exponentially; tobacco heating products (THPs) are less well known but are beginning to establish themselves with consumers in certain markets. Both e-cigarettes and THPs are being offered to the consumer as harm reduction alternatives to cigarette smoking, with these products delivering nicotine via inhaled aerosols with vastly different chemical profiles compared to traditional combusted tobacco. In this study, the aerosol from a new commercially available THP called ***glo***[1] (**FIGURE 1**) was assessed *in vitro*, comparing the cytotoxicity of a continuous lung cell line after exposure to cigarette smoke (3R4F) and *glo* vapour.

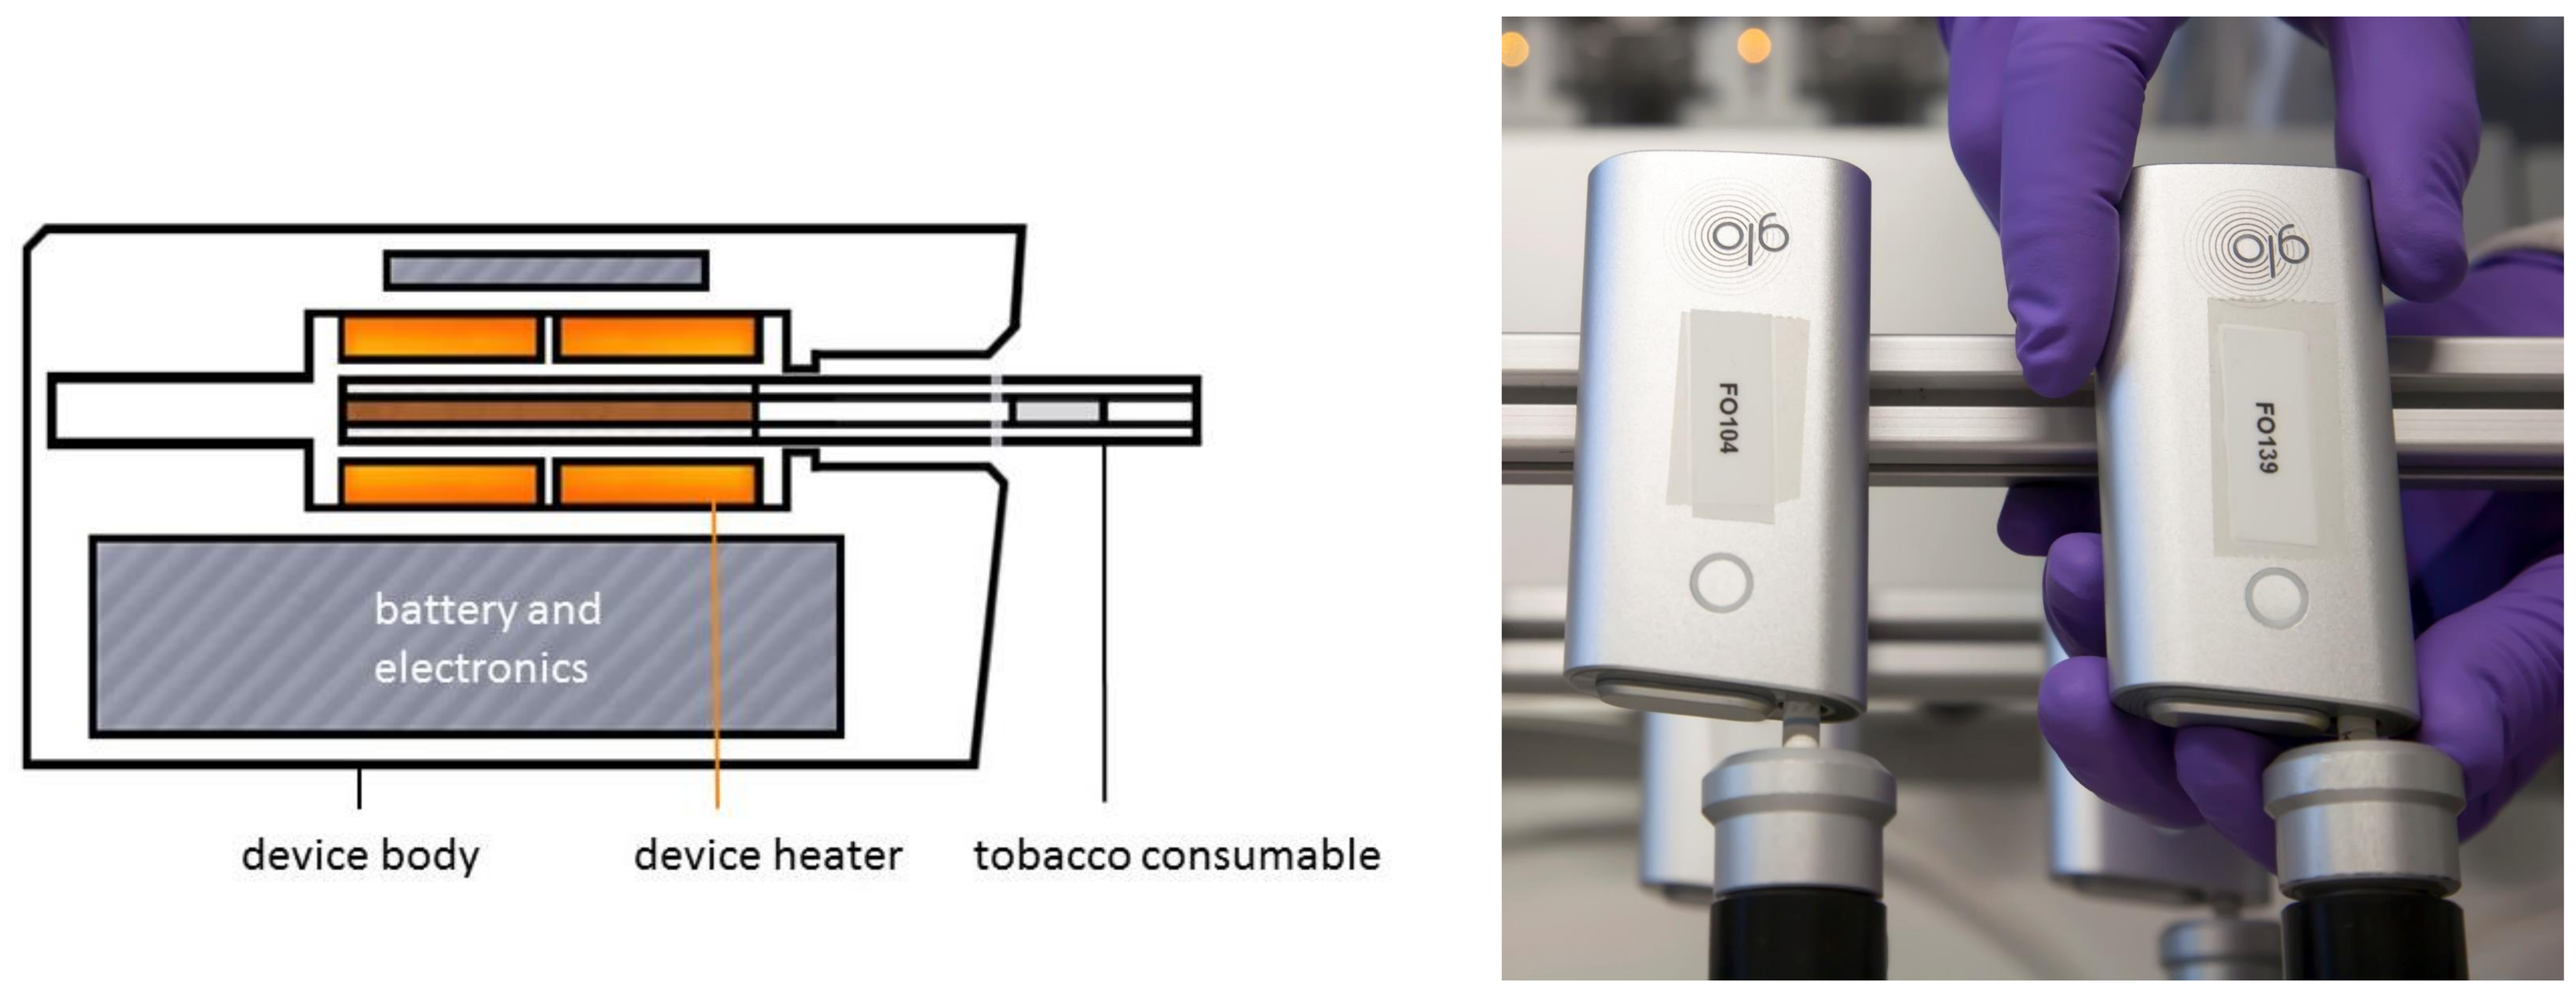
battery and electronics
device body
device heater
tobacco consumable
glo
FO104
glo
FO139

FIGURE 1 A schematic of *glo* (left); *glo* being assessed in the lab (right)

METHODS Exposure to *glo* vapour *in vitro*

A range of biologically relevant aerosol dilutions were generated using the *Borgwaldt RM20S Smoking Machine* (**FIGURE 2A**). The aerosol was delivered to *in vitro* exposure chambers housing H292 lung cell lines at the air liquid interface (**FIGURE 2B**). After a 60-minute exposure (Health Canada smoking regime) lung cells received a 24-hr recovery period. Thereafter, cell viability was assessed by the Neutral Red Uptake assay (NRU)[2].

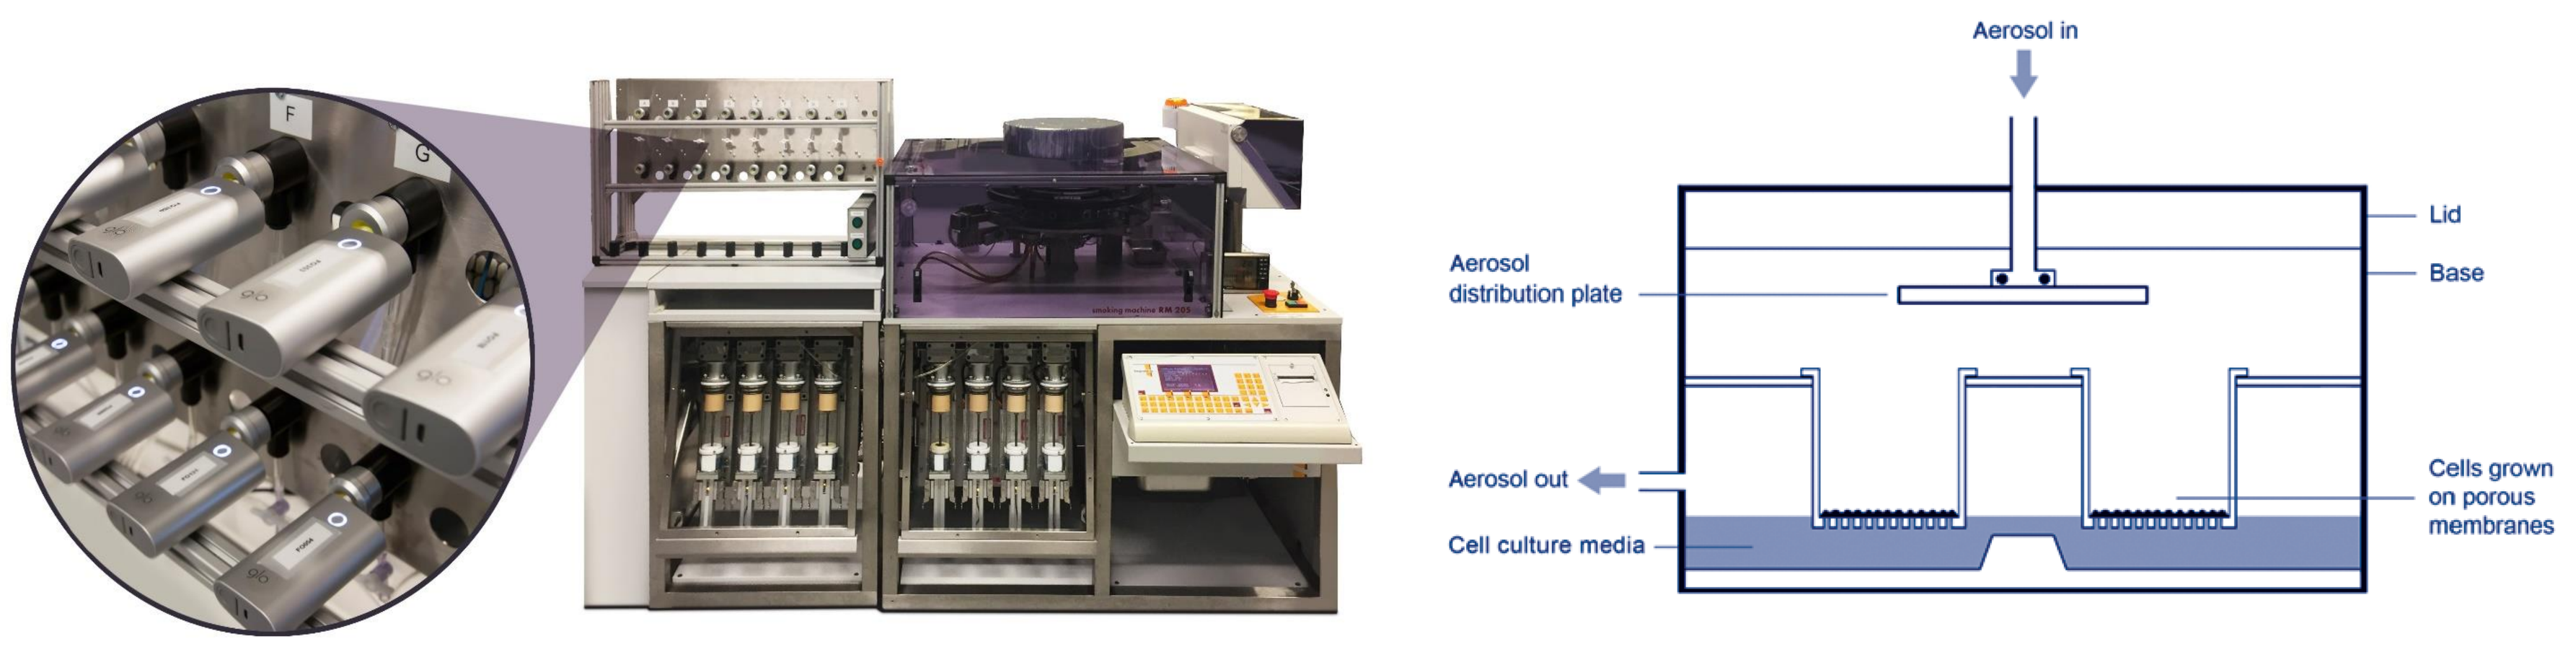
Aerosol in
Lid
Base
Aerosol distribution plate
Aerosol out
Cell culture media
Cells grown on porous membranes

FIGURE 2A Exposure system with *glo*; **2B** *in vitro* exposure chamber

RESULTS Nicotine dosimetry in the exposed culture media

Cell culture media in the base of each exposure chamber (25 ml DMEM) was retained post-exposure for nicotine quantification. This enabled post-exposure cell viability data to be presented as a function of exposed nicotine (a marker common in *glo* vapour and tobacco smoke). Exposed media samples of 1 ml were spiked with d_4-nicotine standard (10 ng/ml) and analysed via liquid chromatography mass spectrometry (UPLC-MS/MS)[3].

Exposed media nicotine concentration was established for each product. As 3R4F cigarette smoke was more concentrated, the biological dilution range selected was greater compared to *glo*, ranging 1:20 – 1:10000 for 3R4F and 1:2 – 1:200 for *glo* (aerosol:air, v:v) (**FIGURE 3**)[4].

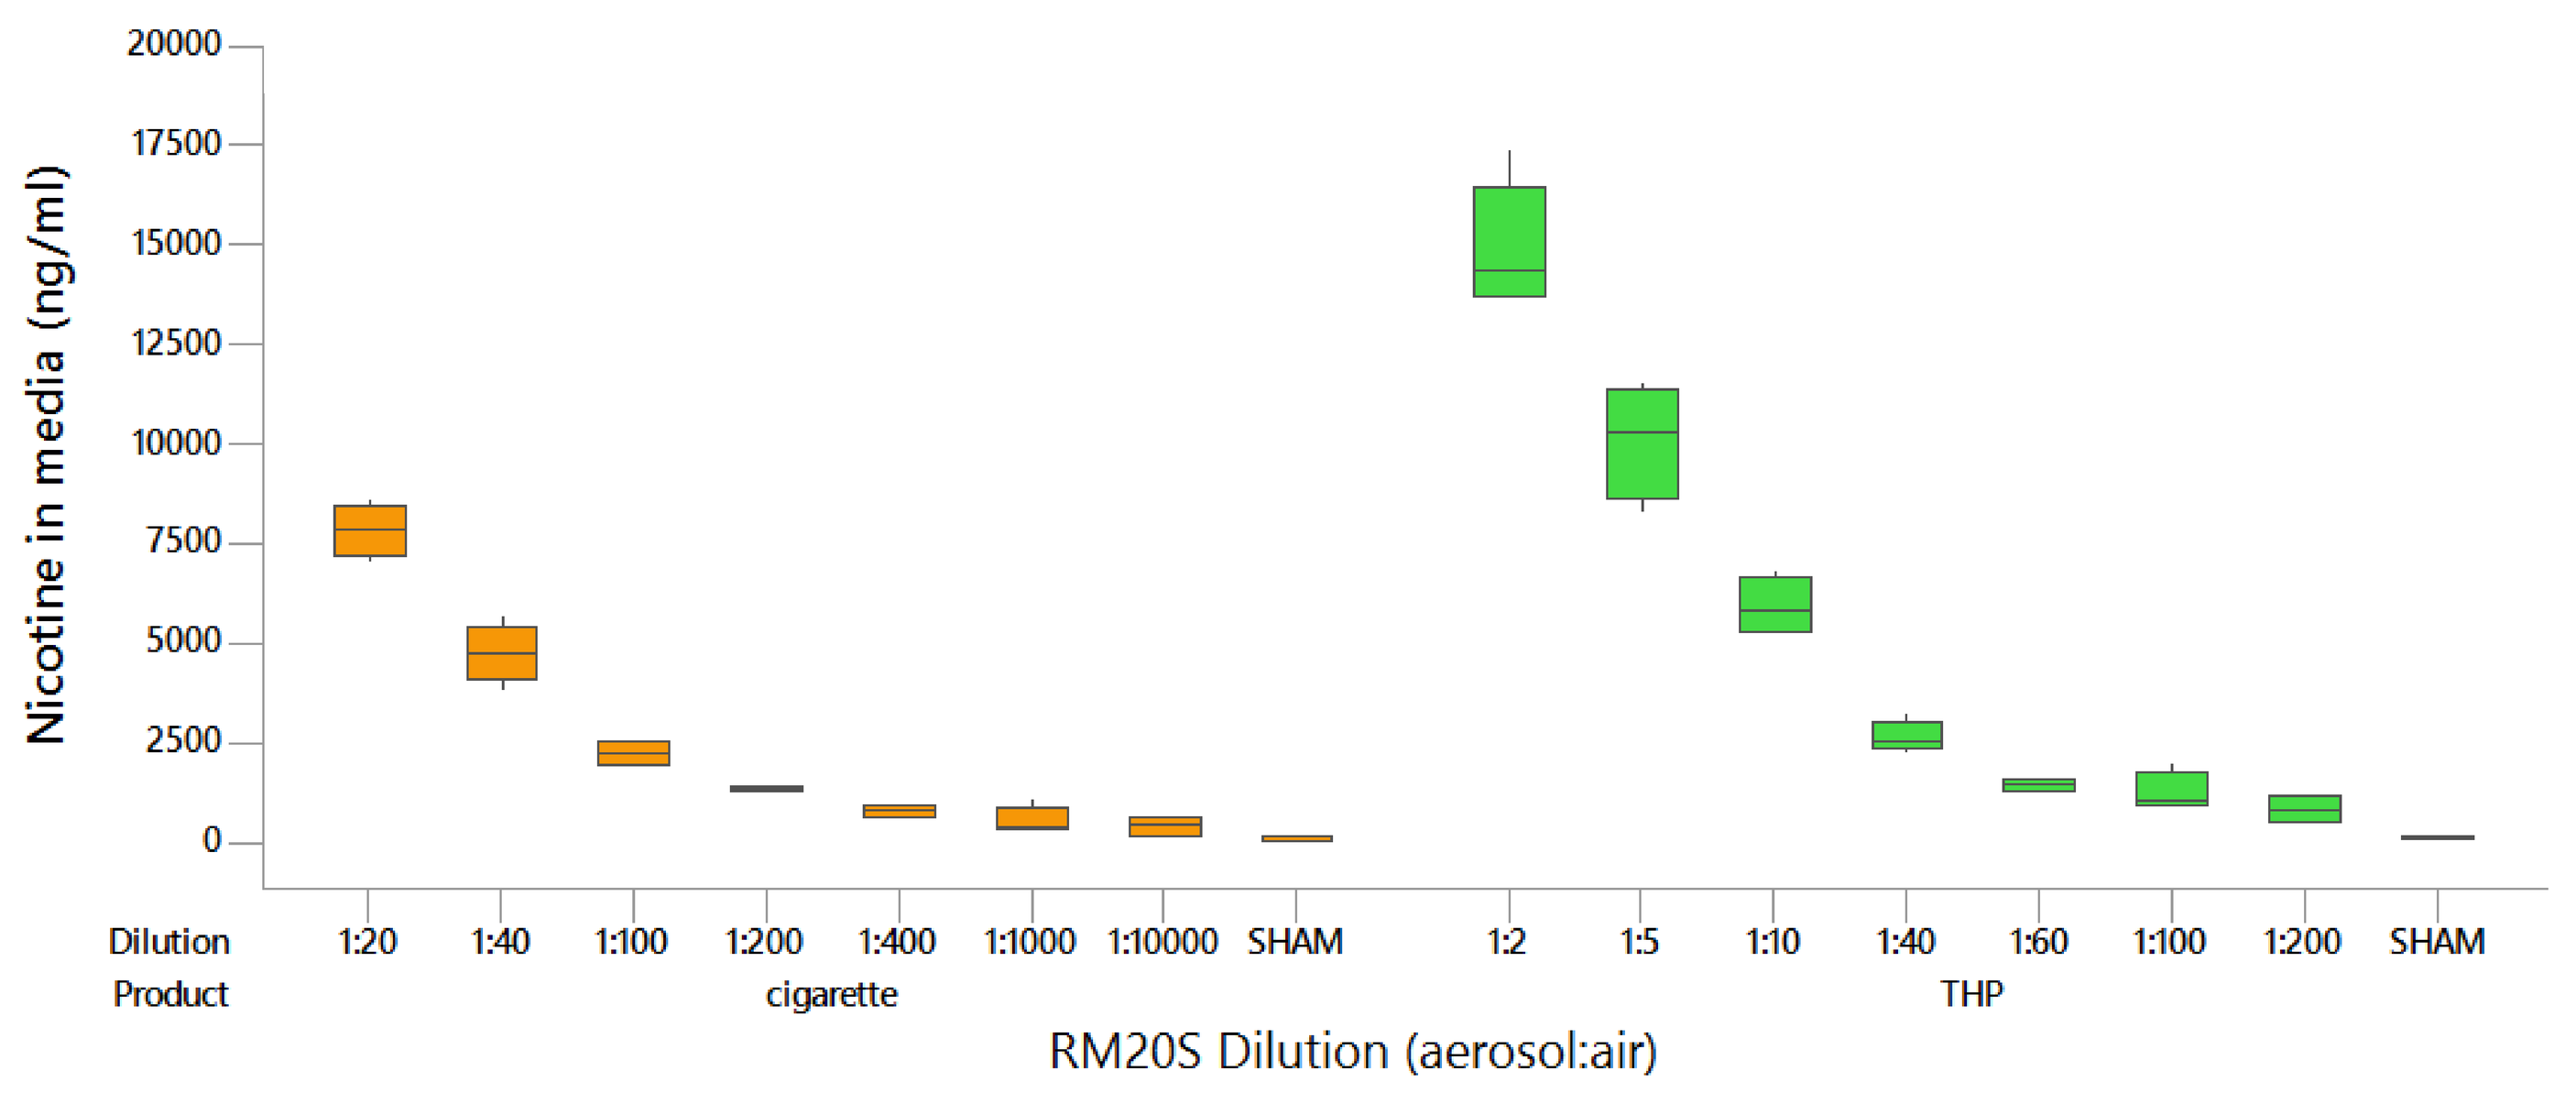
Nicotine in media (ng/ml)
20000
17500
15000
12500
10000
7500
5000
2500
0
Dilution
Product
1:20
1:40
1:100
1:200
1:400
1:1000
1:10000
SHAM
cigarette
1:2
1:5
1:10
1:40
1:60
1:100
1:200
SHAM
THP
RM20S Dilution (aerosol:air)

FIGURE 3 Nicotine dosimetry in the exposed culture media

RESULTS Comparative cytotoxicity

Cytotoxicity was presented against aerosol dilutions (**FIGURE 4A**), and media nicotine concentration (**FIGURE 4B**) to make appropriate exposure comparisons. Cell viability was calculated as a percentage of the air (SHAM) control in each case (n=4/dilution 3R4F; n=7 *glo*). For *glo*, cell viability ranged 92 ± 8% at the highest dilution of vapour at 1:200 to 63 ± 13% at the highest concentration of vapour at 1:2. With the 3R4F smoke, zero percent cell viability (100% cytotoxicity) was reached at a dilution of 1:40 (aerosol:air) and yet at the same dilution, *glo* showed 97% viability[4].

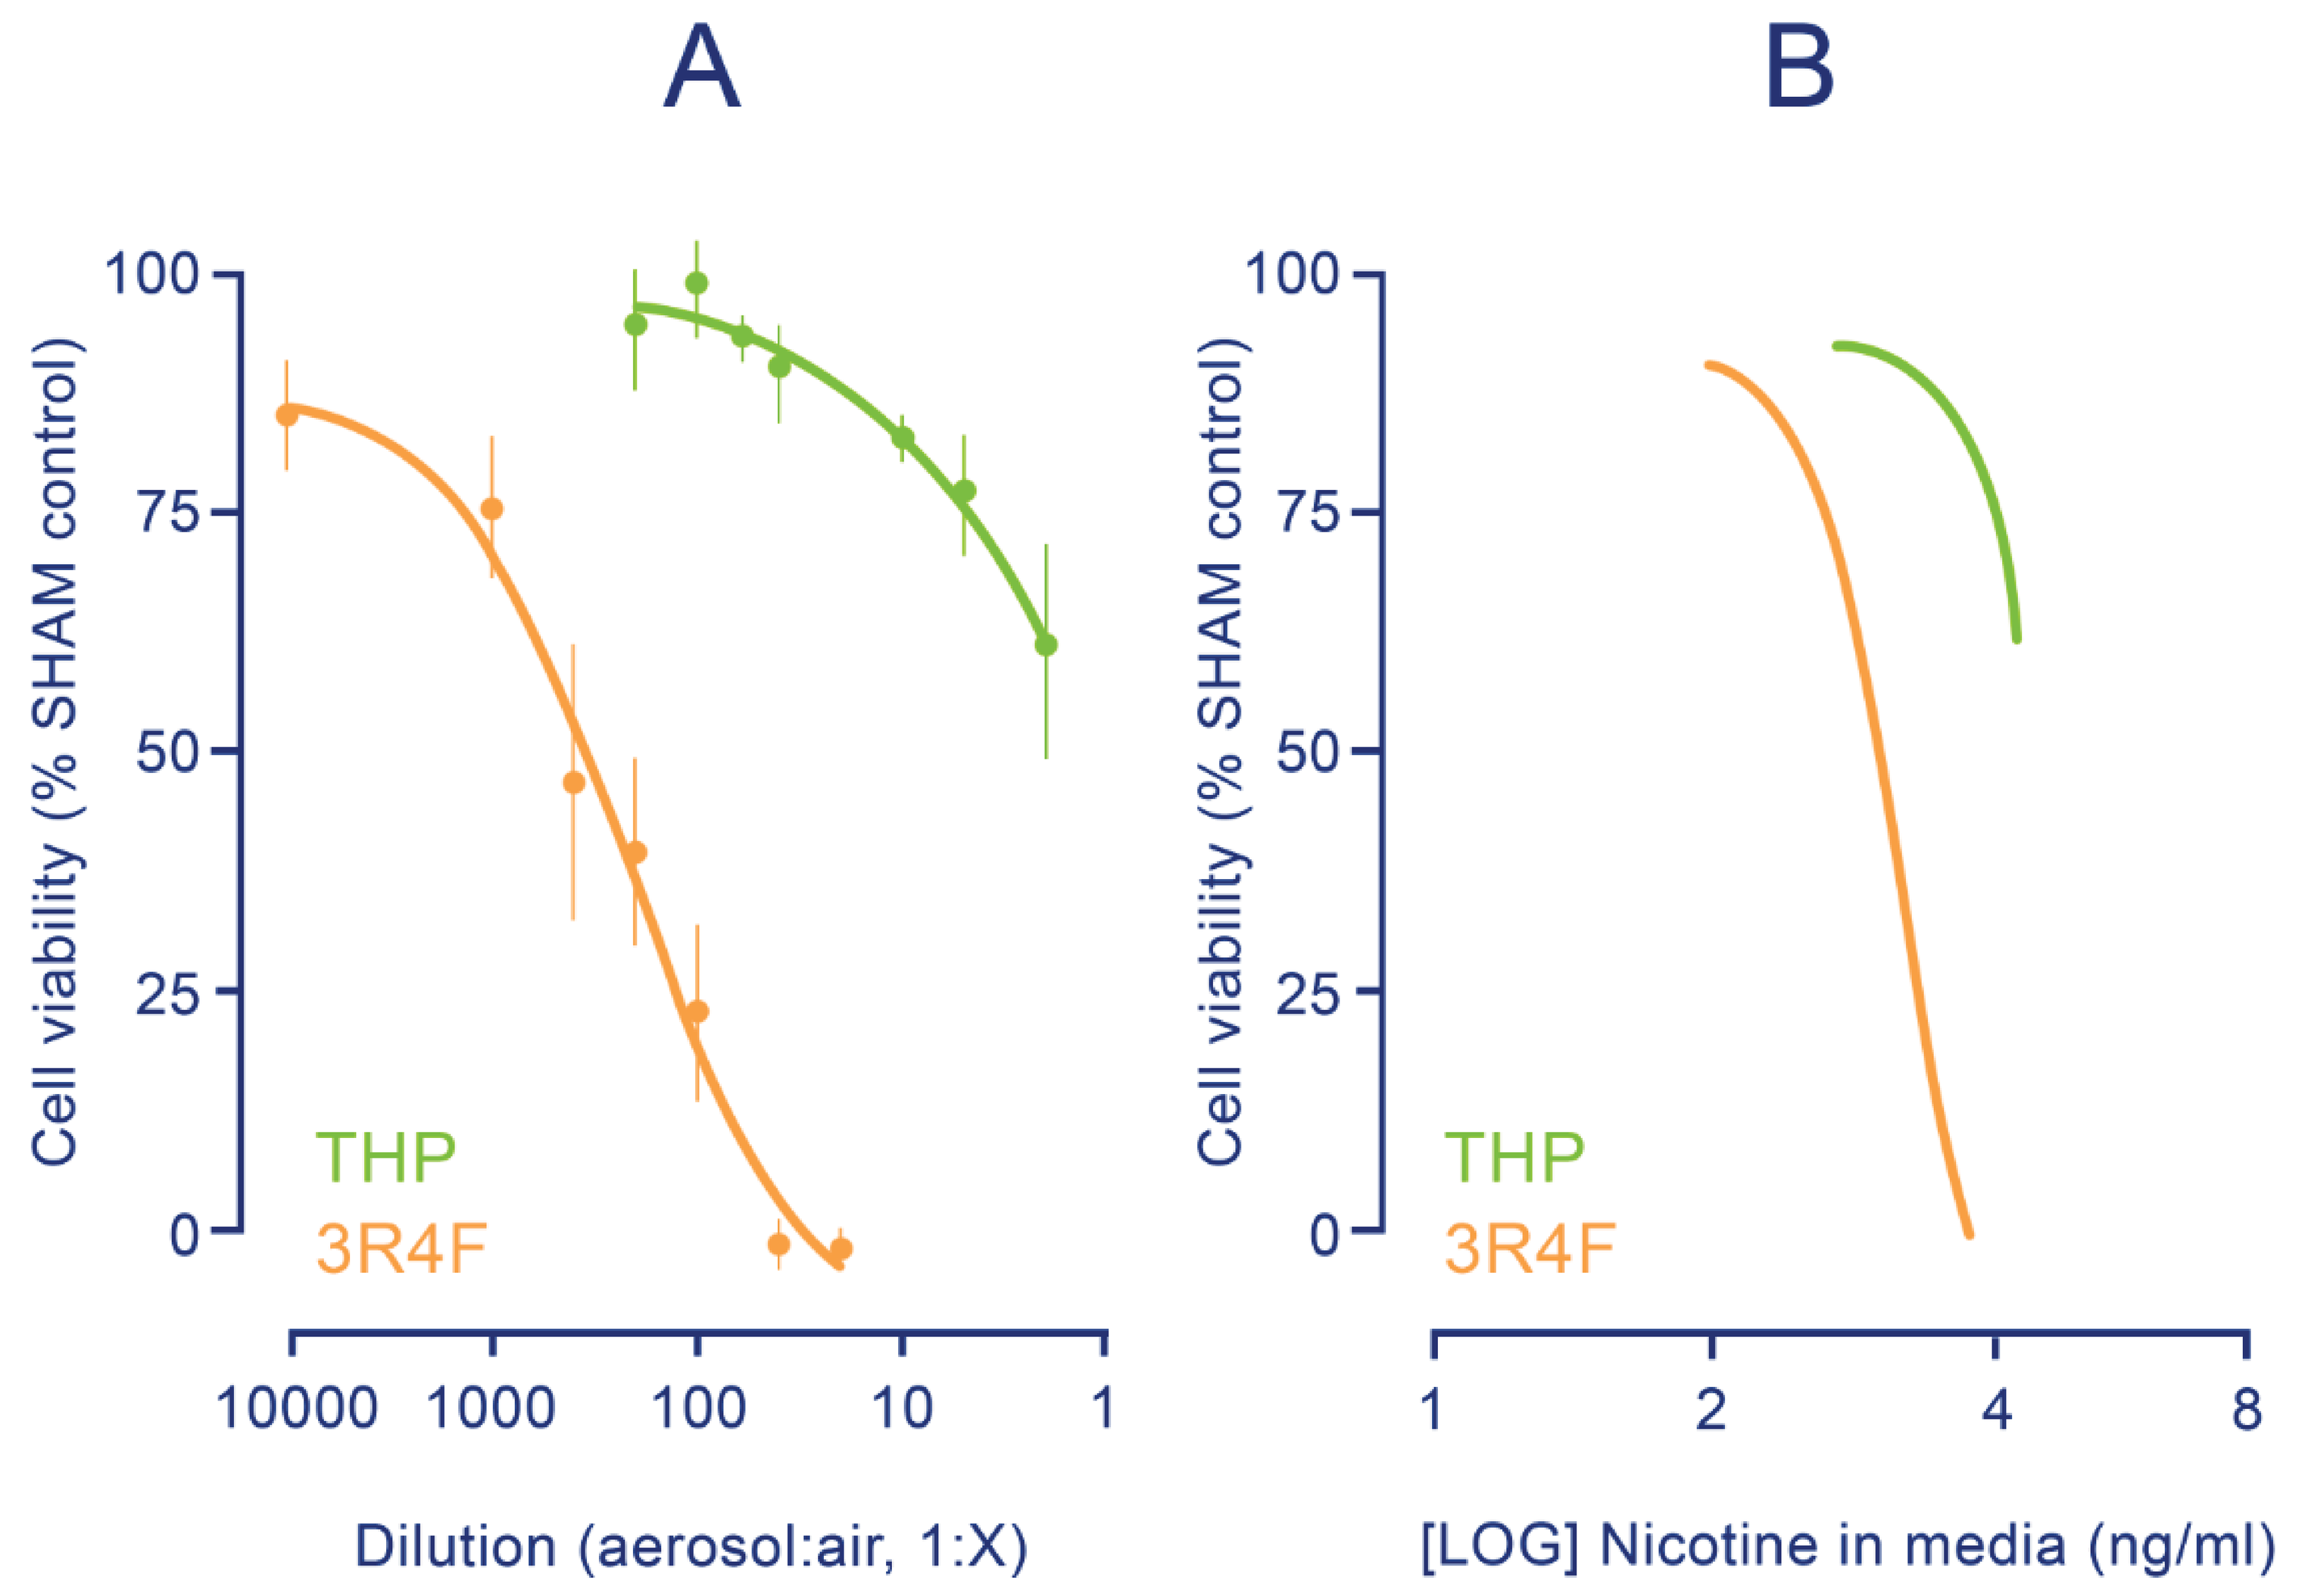
A
B
Cell viability (% SHAM control)
100
75
50
25
0
THP
3R4F
10000
1000
100
10
1
Dilution (aerosol:air, 1:X)
Cell viability (% SHAM control)
100
75
50
25
0
THP
3R4F
1
2
4
8
[LOG] Nicotine in media (ng/ml)

FIGURE 4A Cytotoxicity after 1-hr aerosol exposure versus dilution;
4B Cytotoxicity presented against media nicotine concentration

CONCLUSIONS

Our data demonstrate that the *in vitro* cytotoxic response from a cigarette and THP are significantly different, with the observed frameshift in the THP response clearly indicative of reduced effect. As well as dosimetry data clearly demonstrating cellular exposure, this enables the comparative biological response to be presented against exposed nicotine, and can put such data into wider context of other nicotine delivery products. These *in vitro* data, along with emission chemistries and aerosol characterisation, provide a solid foundation for clinical assessment of our new tobacco heating products.

REFERENCES

1 **Eaton** et al., 2017. Product design, operation and thermophysical characterisation of a novel tobacco heating product, THP1.0(T). Reg. Tox Pharm. In submission
2 **Azzopardi** et al., 2016. Electronic cigarette aerosol induces significantly less cytotoxicity than tobacco smoke. Tox. Mech. and Met. 26:6:477-491
3 **Adamson** et al., 2016. Application of dosimetry tools for the assessment of e-cigarette aerosol and cigarette smoke generated on two different in vitro exposure systems. Chem. Cent. J. 10:74
4 **Jaunky** et al., 2017. Assessment of a novel tobacco heating product THP1.0(T). Part 5: In vitro dosimetric and cytotoxic assessment. Reg. Tox Pharm. In submission.

www.bat-science.com
@BAT_Sci

Poster P3-AH-IND-019
European Aerosol Conference
Zurich, Switzerland **Aug/Sept 2017**